Efficacy Subgroup Analysis: KBP-5074, a Novel Non-Steroidal, Highly Selective, Mineralocorticoid Receptor Antagonist (MRA) for the Treatment of Uncontrolled Hypertension in CKD Stage 3b/4

James McCabe MD, Fred Yang, Vince Benn, Jay Zhang, June Ye, Xiaojuan Tan, Jinrong Liu, Julia Yang, David Hauser

KBP BIOSCIENCES PTE. LTD.

CVCT 2021

ABSTRACT

TITLE:
Subgroup Analysis of the Efficacy of KBP-5074, a Non-Steroidal, Highly Selective, Mineralocorticoid Receptor Antagonist (MRA) in uncontrolled hypertension and CKD Stage 3b/4

BACKGROUND:
KBP-5074, a novel highly protein-bound and highly selective non-steroidal mineralocorticoid receptor antagonist (MRA), is currently under development, for the treatment of uncontrolled hypertension in patients with chronic kidney disease (CKD). KBP-5074 selectively binds to recombinant human mineralocorticoid receptor (MR) with much higher affinity then to recombinant human glucocorticoid, progesterone, and androgen receptors and has a higher antagonistic activity to the MR than steroidal MRAs (ie, spironolactone and eplerenone) and non-steroidal finerenone (1-3). Phase 2b clinical trial BLOCK-CKD (4) with subjects having CKD stage 3b/4 (eGFR 15-44 mL/min/1.73 m^2) demonstrated statistically significant blood pressure lowering and that neither hyperkalemia nor reductions in kidney function were limiting factors to the use of KBP-5074.

OBJECTIVES:
To evaluate the efficacy of KBP-5074 in exploratory subgroups with uncontrolled hypertension in stage 3b/4 CKD.

METHODS:
BLOCK-CKD is a phase 2b, international, multicenter, randomized, double-blind, placebo-controlled, parallel-group study. It evaluates the safety, efficacy, and pharmacokinetics of the nonsteroidal MRA KBP-5074 for uncontrolled hypertension in patients with stage 3b/4 CKD already receiving background antihypertensive medications.

BASELINE CHARACTERISTICS:
A total of 162 patients were randomized. One hundred thirty-eight (138) of 162 (85.2%) patients enrolled completed the study, including 47 patients in the placebo group, 47 patients in the KBP-5074 0.25-mg group, and 44 patients in the KBP-5074 0.50-mg group. Upon the completion of enrollment, 162 stage 3b/4 CKD patients randomized in the clinical trial had a mean SBP at baseline of 155.3 mm Hg, and a DBP of 87.7 mm Hg; 64 (39.5%) patients had stage 4 CKD and 125 (77.2%) patients had proteinuria as defined by UACR ≥30 mg/g and 85 (52.5%) patients having albuminuria (UACR >300 mg/g). Baseline eGFR was similar across treatment groups at baseline was 31.9 mL/min/1.73 m^2. 144 (88.9%) patients were taking at least 3 antihypertensive medications and 81 (50.0%) patients were taking at least 4 antihypertensive medications. A subject was considered to be diabetic if the Investigator determined that diabetes was the etiology of the subject's renal disease; overall, 51 (31.5%) patients were considered diabetic.

RESULTS:
Subgroup analyses for placebo-corrected change from Baseline to Day 84 in SBP resulted in consistent and clinically meaningful reductions from Baseline in SBP (i.e., the placebo-corrected LS mean reduction from Baseline to Day 84 in SBP was >5 mmHg) were observed in subgroups analyzed. In subgroup evaluation of SBP over different age categories (≤65 or >65 years), baseline eGFR (<30 or ≥30 mL/min/1.73 m^2), baseline SBP (≥ 160 vs < 160 mm Hg), baseline antihypertensive medications (3 or more or 2 or less), gender, albuminuria, ethnicity, or diabetic status, consistent results were observed.

CONCLUSIONS:
KBP-5074 demonstrated consistent efficacy across numerous subgroups.

For additional information please contact:

James McCabe MD
Senior Vice President of Clinical Development/Medical Affairs
KBP BIOSCIENCES PTE. LTD.
Email: james.mccabe@kbpbiosciences.com

BACKGROUND

- KBP-5074, a novel highly protein-bound and highly selective non-steroidal mineralocorticoid receptor antagonist (MRA), is currently under development, for the treatment of uncontrolled hypertension in patients with chronic kidney disease (CKD).
- KBP-5074 selectively binds to recombinant human mineralocorticoid receptor (MR) with much higher affinity then to recombinant human glucocorticoid, progesterone, and androgen receptors and has a higher antagonistic activity to the MR than steroidal MRAs (ie, spironolactone and eplerenone) and non-steroidal finerenone (1-3).
- Phase 2b clinical trial BLOCK-CKD (4) with subjects having CKD stage 3b/4 (eGFR 15-44 mL/min/1.73 m^2) demonstrated statistically significant blood pressure lowering and that neither hyperkalemia nor reductions in kidney function were limiting factors to the use of KBP-5074

METHODS

- Overall, of 455 screened subjects, a total of 162 subjects were randomized to either placebo (57 [35.2%] subjects), KBP-5074 0.25-mg (51 [31.5%] subjects) or KBP-5074 0.5-mg (54 [33.3%] subjects) group.
- The randomized subjects were stratified based on their baseline eGFR <30 or > 30 mL/min/1.73 m^2 and SBP <160 or > 160 mm Hg.
- The primary efficacy endpoint was the change in trough cuff seated SBP from Baseline to Day 84.
- Although blood pressure was measured in triplicate, only the average of the last 2 triplicate measurements were used in the analyses.

BASELINE CHARACTERISTICS

- A total of 162 patients were randomized. The baseline demographics and characteristics are shown in Table 1.
- 138 of 162 (85.2%) patients enrolled completed the study, including 47 patients in the placebo group, 47 patients in the KBP-5074 0.25-mg group, and 44 patients in the KBP-5074 0.50-mg group.

Table 1. Baseline Demographics and Characteristics

Baseline Demographics and Characteristics	Overall (N=162)
SBP (mmHg): Mean (SD)	155.3 (13.55)
DBP (mmHg): Mean (SD)	87.7 (12.23)
Background antihypertensive medication: n (%)	
• 3 or more	144 (88.9)
• 2 or less	18 (11.1)
Baseline eGFR (mL/min/1.73 m^2): mean (SD)	31.9 (9.90)
Baseline eGFR: n (%)	
• CKD Stage 3b: 30-44 mL/min/1.73 m^2	98 (60.5)
• CKD Stage 4: 15-29 mL/min/1.73 m^2	64 (39.5)
Baseline Proteinuria: n (%)	
• UACR >300 mg/g	85 (52.5)
• UACR 30-300 mg/g	40 (24.7)
• UACR <30 mg/g	35 (21.6)

PRIMARY EFFICACY ANALYSIS

- The primary analysis showed statistically significant decreases in mean trough cuff seated SBP from Baseline to Day 84 between the KBP-5074 0.25-mg and 0.5-mg groups compared to placebo (Figure 1).
- The decreases in mean trough cuff seated SBP from Baseline to Day 84 were greater in the KBP-5074 0.5-mg (-15.9 mm Hg) and 0.25-mg (-11.5 mm Hg) groups than in the placebo group (-5.3 mm Hg).
- The LS mean treatment differences in change from Baseline to Day 84 between the KBP-5074 0.5-mg and placebo groups (-10.2 mm Hg [95.01% CI: -16.7, -3.6]) and between the KBP-5074 0.25-mg and placebo groups (-7.0 mm Hg [95.01% CI: -13.6, -0.3]) were statistically significant (p=0.0026 and p=0.0399, respectively).

Figure 1. Line Plot of Mean Change in Trough cuff Seated SBP from Baseline to Post-Baseline Visits (LOCF Analysis) (Intent-to-Treat Population)

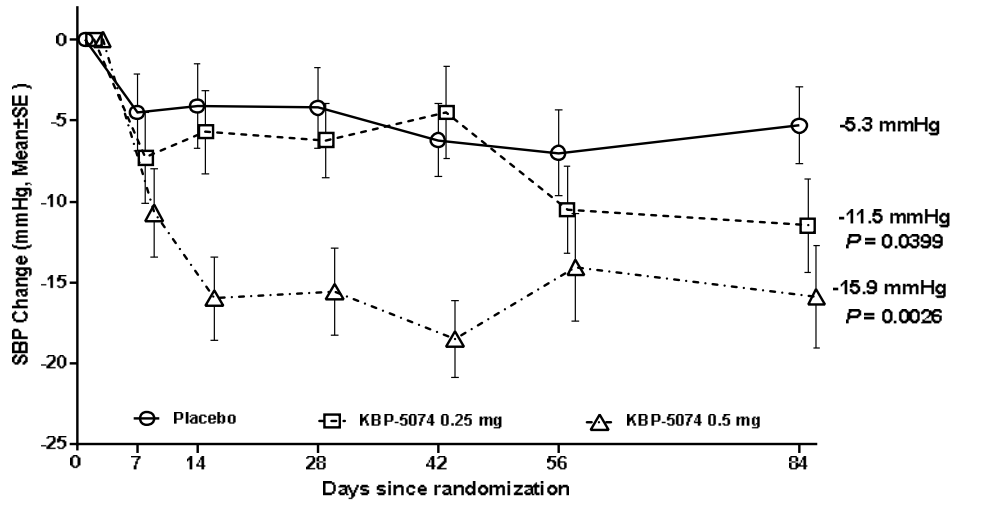
SBP Change (mmHg, Mean±SE)
0
-5
-10
-15
-20
-25
0
7
14
28
42
56
84
Days since randomization
-5.3 mmHg
-11.5 mmHg
P = 0.0399
-15.9 mmHg
P = 0.0026
Placebo
KBP-5074 0.25 mg
KBP-5074 0.5 mg

Note: Baseline was defined as the last non-missing value before the subject received the first dose of study drug.

Abbreviations: SBP, systolic blood pressure; SE, standard error

Source: Table 14.2.1.1

EXPLORATORY SUBGROUP ANALYSES

- Subgroup analyses for placebo-corrected change from Baseline to Day 84 in SBP are shown in Figure 2A and B.
- In general, consistent and clinically meaningful reductions from Baseline in SBP (i.e., the placebo-corrected LS mean reduction from Baseline to Day 84 in SBP was >5 mm Hg) were observed in subgroups analyzed.
- The subgroup analysis showed no differences in reduction from baseline SBP with different age categories (≤65 or >65 years), including group cut off (18 to ≤45 years, >45 to ≤65 years, >65 to ≤75 years, >75 years), baseline eGFR (<30 or ≥30 mL/min/1.73 m^2), baseline SBP (≥160 vs < 160 mm Hg), baseline antihypertensive medications (3 or more or 2 or less), gender, ethnicity, or diabetic status, consistent results were observed.
- KBP-5074 resulted in a consistent, dose-dependent and statistically significant lowering of SBP in subjects with albuminuria (UACR >300 mg/g). In subjects with UACR <30 mg/g, a dose-dependent trend towards lower SBP was observed with statistically significant lowering of SBP in the KBP-5074 0.5-mg group compared to placebo (p=0.0301).

Figure 2. Subgroup analyses for placebo-corrected mean change from Baseline to Day 84 in SBP for KBP-5074 treatment groups (Intent-to-Treat Population)

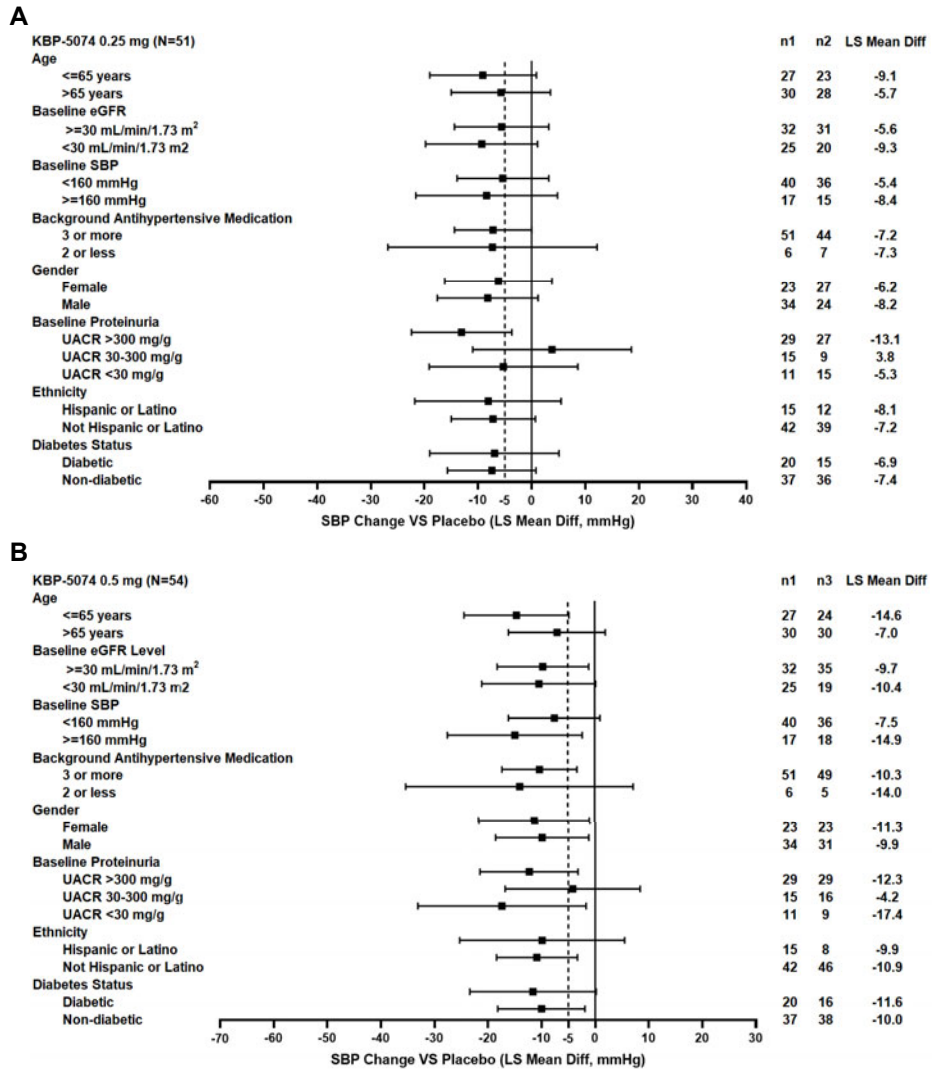
A
KBP-5074 0.25 mg (N=51)
n1 n2 LS Mean Diff
Age
<=65 years 27 23 -9.1
>65 years 30 28 -5.7
Baseline eGFR
>=30 mL/min/1.73 m² 32 31 -5.6
<30 mL/min/1.73 m2 25 20 -9.3
Baseline SBP
<160 mmHg 40 36 -5.4
>=160 mmHg 17 15 -8.4
Background Antihypertensive Medication
3 or more 51 44 -7.2
2 or less 6 7 -7.3
Gender
Female 23 27 -6.2
Male 34 24 -8.2
Baseline Proteinuria
UACR >300 mg/g 29 27 -13.1
UACR 30-300 mg/g 15 9 3.8
UACR <30 mg/g 11 15 -5.3
Ethnicity
Hispanic or Latino 15 12 -8.1
Not Hispanic or Latino 42 39 -7.2
Diabetes Status
Diabetic 20 15 -6.9
Non-diabetic 37 36 -7.4
-60 -50 -40 -30 -20 -10 -5 0 10 20 30 40
SBP Change VS Placebo (LS Mean Diff, mmHg)
B
KBP-5074 0.5 mg (N=54)
n1 n3 LS Mean Diff
Age
<=65 years 27 24 -14.6
>65 years 30 30 -7.0
Baseline eGFR Level
>=30 mL/min/1.73 m² 32 35 -9.7
<30 mL/min/1.73 m2 25 19 -10.4
Baseline SBP
<160 mmHg 40 36 -7.5
>=160 mmHg 17 18 -14.9
Background Antihypertensive Medication
3 or more 51 49 -10.3
2 or less 6 5 -14.0
Gender
Female 23 23 -11.3
Male 34 31 -9.9
Baseline Proteinuria
UACR >300 mg/g 29 29 -12.3
UACR 30-300 mg/g 15 16 -4.2
UACR <30 mg/g 11 9 -17.4
Ethnicity
Hispanic or Latino 15 8 -9.9
Not Hispanic or Latino 42 46 -10.9
Diabetes Status
Diabetic 20 16 -11.6
Non-diabetic 37 38 -10.0
-70 -60 -50 -40 -30 -20 -10 -5 0 10 20 30
SBP Change VS Placebo (LS Mean Diff, mmHg)

Abbreviations: eGFR, estimated glomerular filtration rate; LS Mean Diff, least squares mean difference; n1, number of subjects on placebo; n2, number of subjects on KBP-5074 0.25-mg QD; n3, number of subjects on KBP-5074 0.5-mg QD; SBP, systolic blood pressure; UACR, urinary albumin creatinine ratio; VS, versus

Note: The dotted line indicates a 5 mm Hg placebo-corrected reduction from Baseline.

Source: Table 14.2.1.7b, Table 14.2.1.8, Table 14.2.1.9, Table 14.2.1.10, Table 14.2.1.12, Table 14.2.1.13 and Table 14.2.1.14

CONCLUSIONS

- This study showed clinically meaningful and statistically significant effects on primary outcome, decreases in trough cuff seated SBP from Baseline to Day 84, for both doses of KBP-5074 compared to placebo in subjects with advanced CKD (eGFR ≥15 to ≤44 mL/min/1.73 m^2) and uncontrolled hypertension (average baseline 155.3 mm Hg) even with multiple background antihypertensive medications (88.9% of subjects on at least 3 antihypertensive medications).
- Subgroup analysis showed consistent SBP reduction across various subgroups such as age, baseline eGFR, baseline SBP, baseline antihypertensive medications, gender, albuminuria, ethnicity, or diabetic status.

References

1. Chow C, Liu J, and Tan X, et al. Pharmacological profile of KBP-5074, a novel non-steroidal mineralocorticoid receptor antagonist for the treatment of cardiorenal diseases. Journal of Drug Research and Development. 2017;3(2):ISSN 2470-1009.
2. Pitt B, Filippatos G, Gheorghiade M, et al. Rationale and design of ARTS: a randomized, double-blind study of BAY 94-8862 in patients with chronic heart failure and mild or moderate chronic kidney disease. Eur J Heart Fail. 2012;14(6):668–675.
3. Pitt B, Jaisser F, Bakris GL. An evaluation of KBP-5074 in advanced chronic kidney disease with uncontrolled hypertension. Expert Opinion on Investigational Drugs. 2021;30(10):1017–1023.
4. Bakris G, Pergola PE, and Delgado B, et al. Effect of KBP-5074 on Blood Pressure in Advanced Chronic Kidney Disease: Results of the BLOCK-CKD Study. Hypertension. 2021;78:74–81.